BIO-BASED PLASTICS FOR MEDICAL DEVICES: A CASE STUDY

Philip Smith, Vectura

INTRODUCTION

Drug delivery devices and particularly those designed for home use and user convenience make great use of the benefits of injection molded plastic in many of their constituent components. For Vectura, and its range of high production volume Dry Powder Inhalers (DPIs), this is no different.

This paper presents the alternatives and tracks the application of Bio-based plastics through to acceptance for use in a proprietary Vectura DPI, the Open Inhale Close (OIC) device, considering the regulatory challenge posed in effecting this change. The use of injection molded plastics allows for repeatable part complexity at scale with well-established processes and with engineered plastics that provide good functional properties as well as meeting and maintaining patient safety.

At the end of their life, these devices are often difficult to manage due to drug contamination and the single use nature rooted in their development. It is particularly true for commercialized or soon to be commercialized devices. Re-design is not viable and the impact on the environment today is greatest.

It is difficult for an individual device manufacturer to influence the wider plastics industry, dominated as it is by non-pharmaceutical industries and reliant on fossil fuels.

This paper presents the alternatives and tracks the application of Bio-based plastics through to acceptance for use in a proprietary Vectura DPI, the Open Inhale Close (OIC) device, considering the regulatory challenge posed in effecting this change.

SUSTAINABLE OPTIONS

Considering that we cannot move away from injection molded plastics in the short term, the options for more sustainable plastics are thus:

Conventional Recycling

Products that contain post-industrial or post-consumer waste. Proving the safety of these plastics is difficult and the variability in quality challenging.
Not Suitable for medical applications

Bio-based Plastics

Made or derived from naturally occurring products, green feedstocks, instead of fossil feedstocks. Commercially available solutions for OIC [2-4].
Suitable for medical applications

Closed Loop Recycling

Returning molded scrap within the supply chain to be reused. Regrind, where excess material, is fedback into molding, is not permitted.
Not Suitable for medical applications

Chemical Recycling

Products which are of virgin quality but produced with feedstocks derived from chemical recycling. This is not industrialized. [1]
Potential for medical applications

RDD 2024 | MAY 5-9, 2024 | TUCSON, AZ, USA

BIO-BASED PLASTICS FOR MEDICAL DEVICES

By mixing equivalent feedstocks to fossil-based materials with those derived from bio-based sources during polymer manufacture, and accounting for this in the output, identical Bio-based and Fossil based polymers can be produced. Figure 1 provides an overview of the production process for medical device plastics [5].

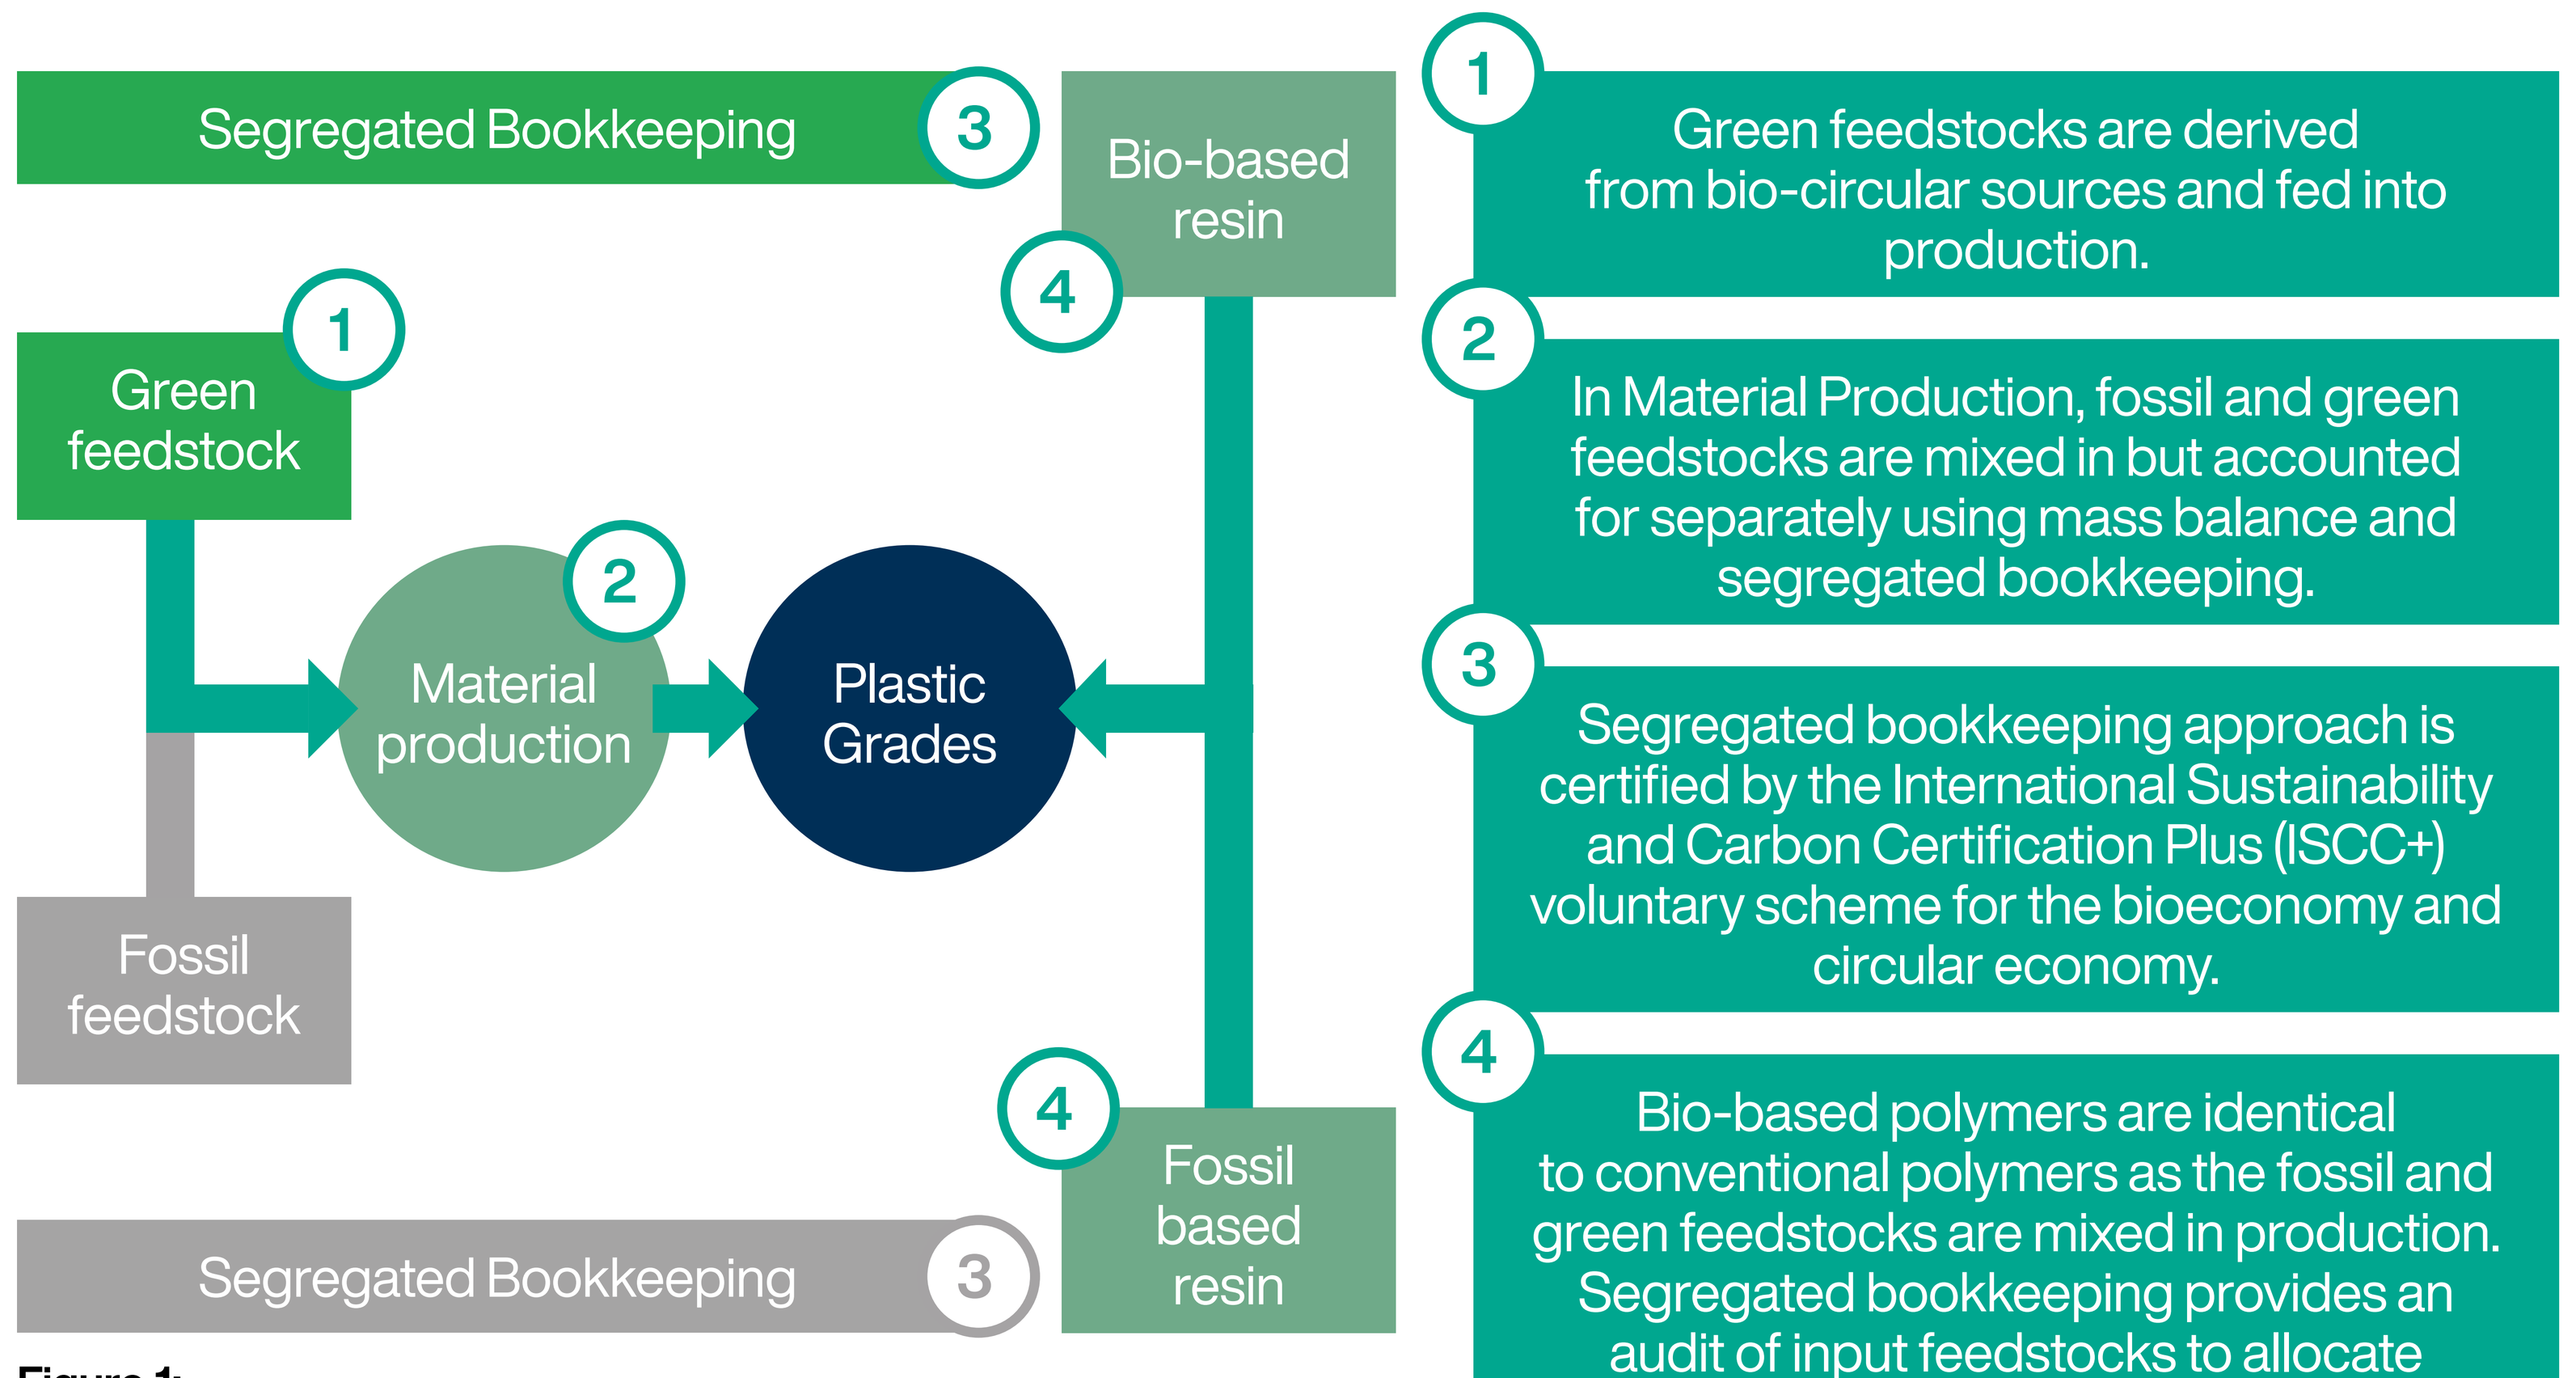
Segregated Bookkeeping
3
Bio-based resin
4
Green feedstock
1
2
Material production
Plastic Grades
Fossil feedstock
4
Fossil based resin
Segregated Bookkeeping
3
1
Green feedstocks are derived from bio-circular sources and fed into production.
2
In Material Production, fossil and green feedstocks are mixed in but accounted for separately using mass balance and segregated bookkeeping.
3
Segregated bookkeeping approach is certified by the International Sustainability and Carbon Certification Plus (ISCC+) voluntary scheme for the bioeconomy and circular economy.
4
Bio-based polymers are identical to conventional polymers as the fossil and green feedstocks are mixed in production. Segregated bookkeeping provides an audit of input feedstocks to allocate production output.

Figure 1:
Medical device plastics production incorporating Bio-based feedstocks.

Bio-based polymers are available for many commonly used plastics in medical devices. In the case of Vectura's Open Inhale Close (OIC) device, all injection molded plastics have a bio-based equivalent which can be used as a drop in, provided the risk of the change can be adequately resolved. The risks pertain to the successful completion of Design Verification and Validation and achieving adequate material compatibility with the target API.

ENVIRONMENTAL IMPACT

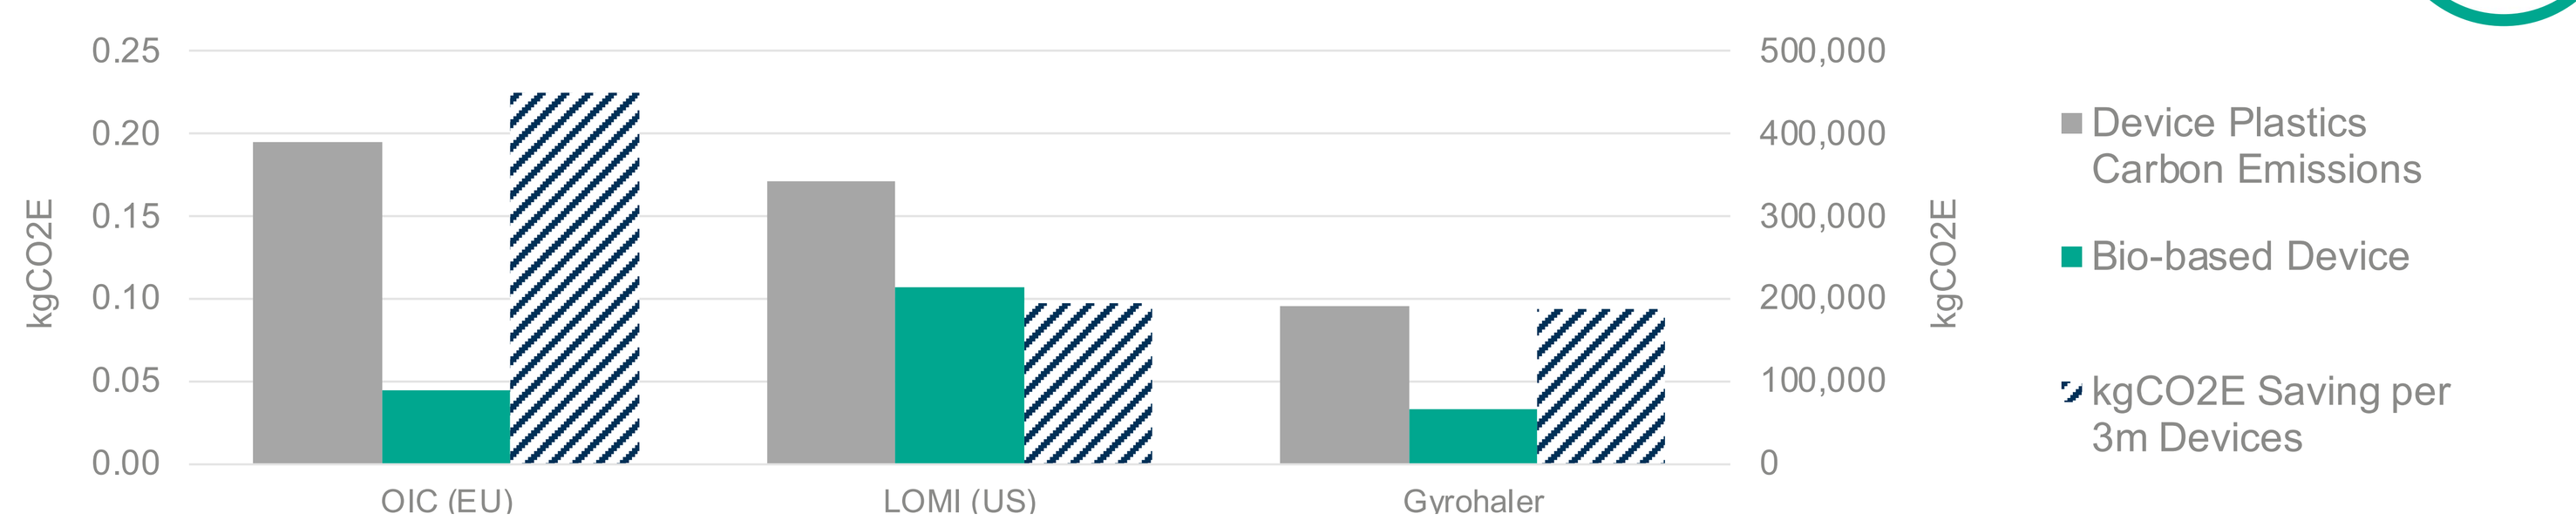
0.25
0.20
0.15
0.10
0.05
0.00
kgCO2E
500,000
400,000
300,000
200,000
100,000
0
kgCO2E
OIC (EU)
LOMI (US)
Gyrohaler
Device Plastics Carbon Emissions
Bio-based Device
kgCO2E Saving per 3m Devices

The benefits of bio-based plastics on sustainability and the environment derive from reducing the need to extract and refine virgin raw materials. Typically, these are fossil in origin, but may extend to other mined substances.

Using bio-circular sources reduces the negative externalities associated with plant-based plastics, such as food crop production and biodiversity loss from land use.

There is a decrease in the life cycle carbon dioxide emissions of Bio-based plastics because they typically use bio-circular sources. Figure 2 compares the emissions from device plastics supply for three of Vectura's key Drug Delivery Devices using data from material suppliers. By isolating this carbon footprint to the raw plastic material elements only allows actual carbon emission reduction to be derived for a typical annual production of 3 million devices, though does not consider the full life cycle of the device [6].

Further benefits include the localization of feedstock production. Bio-circular sources are typically universal, meaning the supply chain does not rely on geopolitics and the associated supply tensions placed by the global distribution of natural resources.

IMPACT TO THE DEVICE

Documentation defining material properties and accreditations along with ISCC+ certification from the material suppliers is reviewed against device material requirements and provides the evidence that the bio-based material is viable.

Whilst the bio-based materials are fundamentally identical to the conventional materials and should provide no risk to device function or patient safety, according to guidance [7], they would still be treated as a material change from a Regulator perspective, and therefore the associated regulatory risks and impacts must be considered.

Changing the device design definition to Bio-based plastics before device Verification and Validation (V&V), diminishes these risks. For the OIC, V&V includes bio-compatibility testing with the target API, which sets material choices, and had not been started prior to the selection of bio-based materials. Subsequent proprietary projects using the OIC device platform with Bio-based materials would only need to repeat V&V activities associated with a change of target API.

There is a cost impact to the approach, and questions of supply chain resilience to consider. However, the cost impact is approximately 1% at a product level. Similarly, supply chain risks are either identical to fossil grades due to co-production or improved by the distribution of feedstock production.

CONCLUSIONS AND NEXT STEPS

Short term steps to improve the sustainability of medical devices can be taken now. The implementation of bio-based plastics is best done at the early stages of development, but slot in solutions, where available, allow Bio-based material selection to be utilized at mold tool qualification, just prior to Design Verification and Validation in the development cycle.

The question of applicability to marketed devices is an open one, and where next steps in the approach to regulation are required. Understanding among all stakeholders is needed to allow the considerable impact of legacy plastics supply to be reduced using Bio-based plastics, and concerted long term change is needed across the industry to minimize the impact of single use plastics at end of life.

REFERENCES

1. Plastics Europe, Chemical recycling and the role of mass balance explained, 2023
2. Covestro showcases Makrolon® RE polycarbonate for healthcare [https://www.covestro.com/en/company/covestro-worldwide/united-states/media/makrolon-re-polycarbonate-for-healthcare], Accessed February 13, 2024.
3. Celanese launches POM ECO-B as mass-balance bio-based option for existing products [https://www.celanese.com/news-and-media/2020/November/celanese-launches-pom-eco-b], Accessed February 13, 2024.
4. Ineos Styrolution recycled and bio-attributed styrenics [https://styrolution-eco.com/eco-products.html], Accessed February 13, 2024.
5. Ellen MacArthur Foundation, Enabling a circular economy for chemicals with the mass balance approach [https://www.ellenmacarthurfoundation.org/white-papers-and-articles], Accessed February 13, 2024
6. Willoughby A, "Assessing the Environmental Impact of Global Supply Chain Logistics and Supplier Selection". ONdrugDelivery 2022, 139: 30-34
7. FDA Guidance (2017): Guidance for Industry and Food and Drug Administration Staff. Deciding When to Submit a 510(k) for a Change to an Existing Device.

Scan QR code to download this poster